02-020

A PHASE 1/2 OPEN-LABEL STUDY TO INVESTIGATE THE SAFETY OF TRANSPLANTATION(by injection) OF HUMAN GLIAL RESTRICTED PROGENITOR CELLS INTO SUBJECTS WITH TRANSVERSE MYELITIS

UTSouthwestern
O'Donnell Brain Institute

JAN CAMERON WATTS, R.N., CARLOS BAGLEY, M.D., SONIA GARG,M.D., BENJAMIN GREENBERG, M.D, MHS.

BACKGROUND

Transverse Myelitis (TM) is an inflammatory condition of the spinal cord that leads to demyelination in the ascending sensory and descending motor tracts. TM affects roughly 1,400 people in the U.S. per year and can cause sensory loss, weakness, bowel and bladder dysfunction. TM affects individuals of all ages, with bimodal peaks between the ages of 10 and 19 years and 30 and 39 years. There is no sex or familial predisposition to TM. When the maximal level of deficit is reached, approximately 50% of patients have lost all movements of their legs, virtually all patients have bladder dysfunction, and 80 – 94% of patients have numbness, paresthesias, or band-like dysesthesias.

OBJECTIVE

- The primary objective of the study is to evaluate the safety of oligoprogenitor Q-Cells® transplantation into the posterior columns of the spinal cord in patients with Transverse Myelitis
- A secondary objective of the study is to obtain preliminary data regarding the clinical activity of Q-Cells® in patients with TM.

DESIGN

- Enrollment is planned for up 9 subjects over 12 months into a phase 1 study. The study will include 3 cohorts receiving escalating 'doses' of cells. Each subject will receive a single time point administration of Q-Cells® with 10 unilateral injections targeted to posterior columns of the spinal cord.
- Study participants will be assessed for safety and potential therapeutic activity.
- Involvement consists of Screening, Pre-operative/Treatment, and Post-treatment study periods. Long term follow up for long term clinical and MRI safety assessments are planned.
- Subjects must have the ability to undergo a laminectomy, and immunosuppressive therapy for a prescribed period. Subjects must be non-ambulatory and able to complete MRI's, and complete multiple follow-up visits.

INCLUSION CRITERIA

- Subject is 18 - 70 years of age (inclusive) on day of Screening Visit.
- Subject more than 12 months out from TM onset
- Subject is diagnosed idiopathic TM within the past 120 months.
- Ability to understand the purpose and risks of the study and provide signed and dated informed consent and authorization to collect and use protected health information (PHI) in accordance with national and local subject privacy regulations.
- Live within reasonable travel distance to center or have a reliable mechanism to travel to the center.
- Have a caregiver willing/able to assist in the transportation and care required by study participation.
- Subject has a MRI with a single focus of T2 hyperintensity that is 4 to 10 cm in length if no post contrast enhancement seen, or a single focus T1 post contrast enhancing lesion of 4 to 10 cm, located below C7
- Subject has negative NMO IgG test at two separate time points, separated by at least 6 months
- Subject has a brain MRI not consistent with multiple sclerosis
- Subject is more than 12 months from TM onset
- Subject is not ambulatory.
- Subject's deficits have been stable for at least 3 months
- Subject is medically able to undergo the study procedures and physically able to adhere to the visit schedule at the time of study entry.
- For women of child bearing capacity, a negative serum pregnancy test during the Screening Period and at the Pre-Operative Visit.
- Males and females should agree to practice effective birth control during study participation.

BOULIS STABLIZATION DEVICE

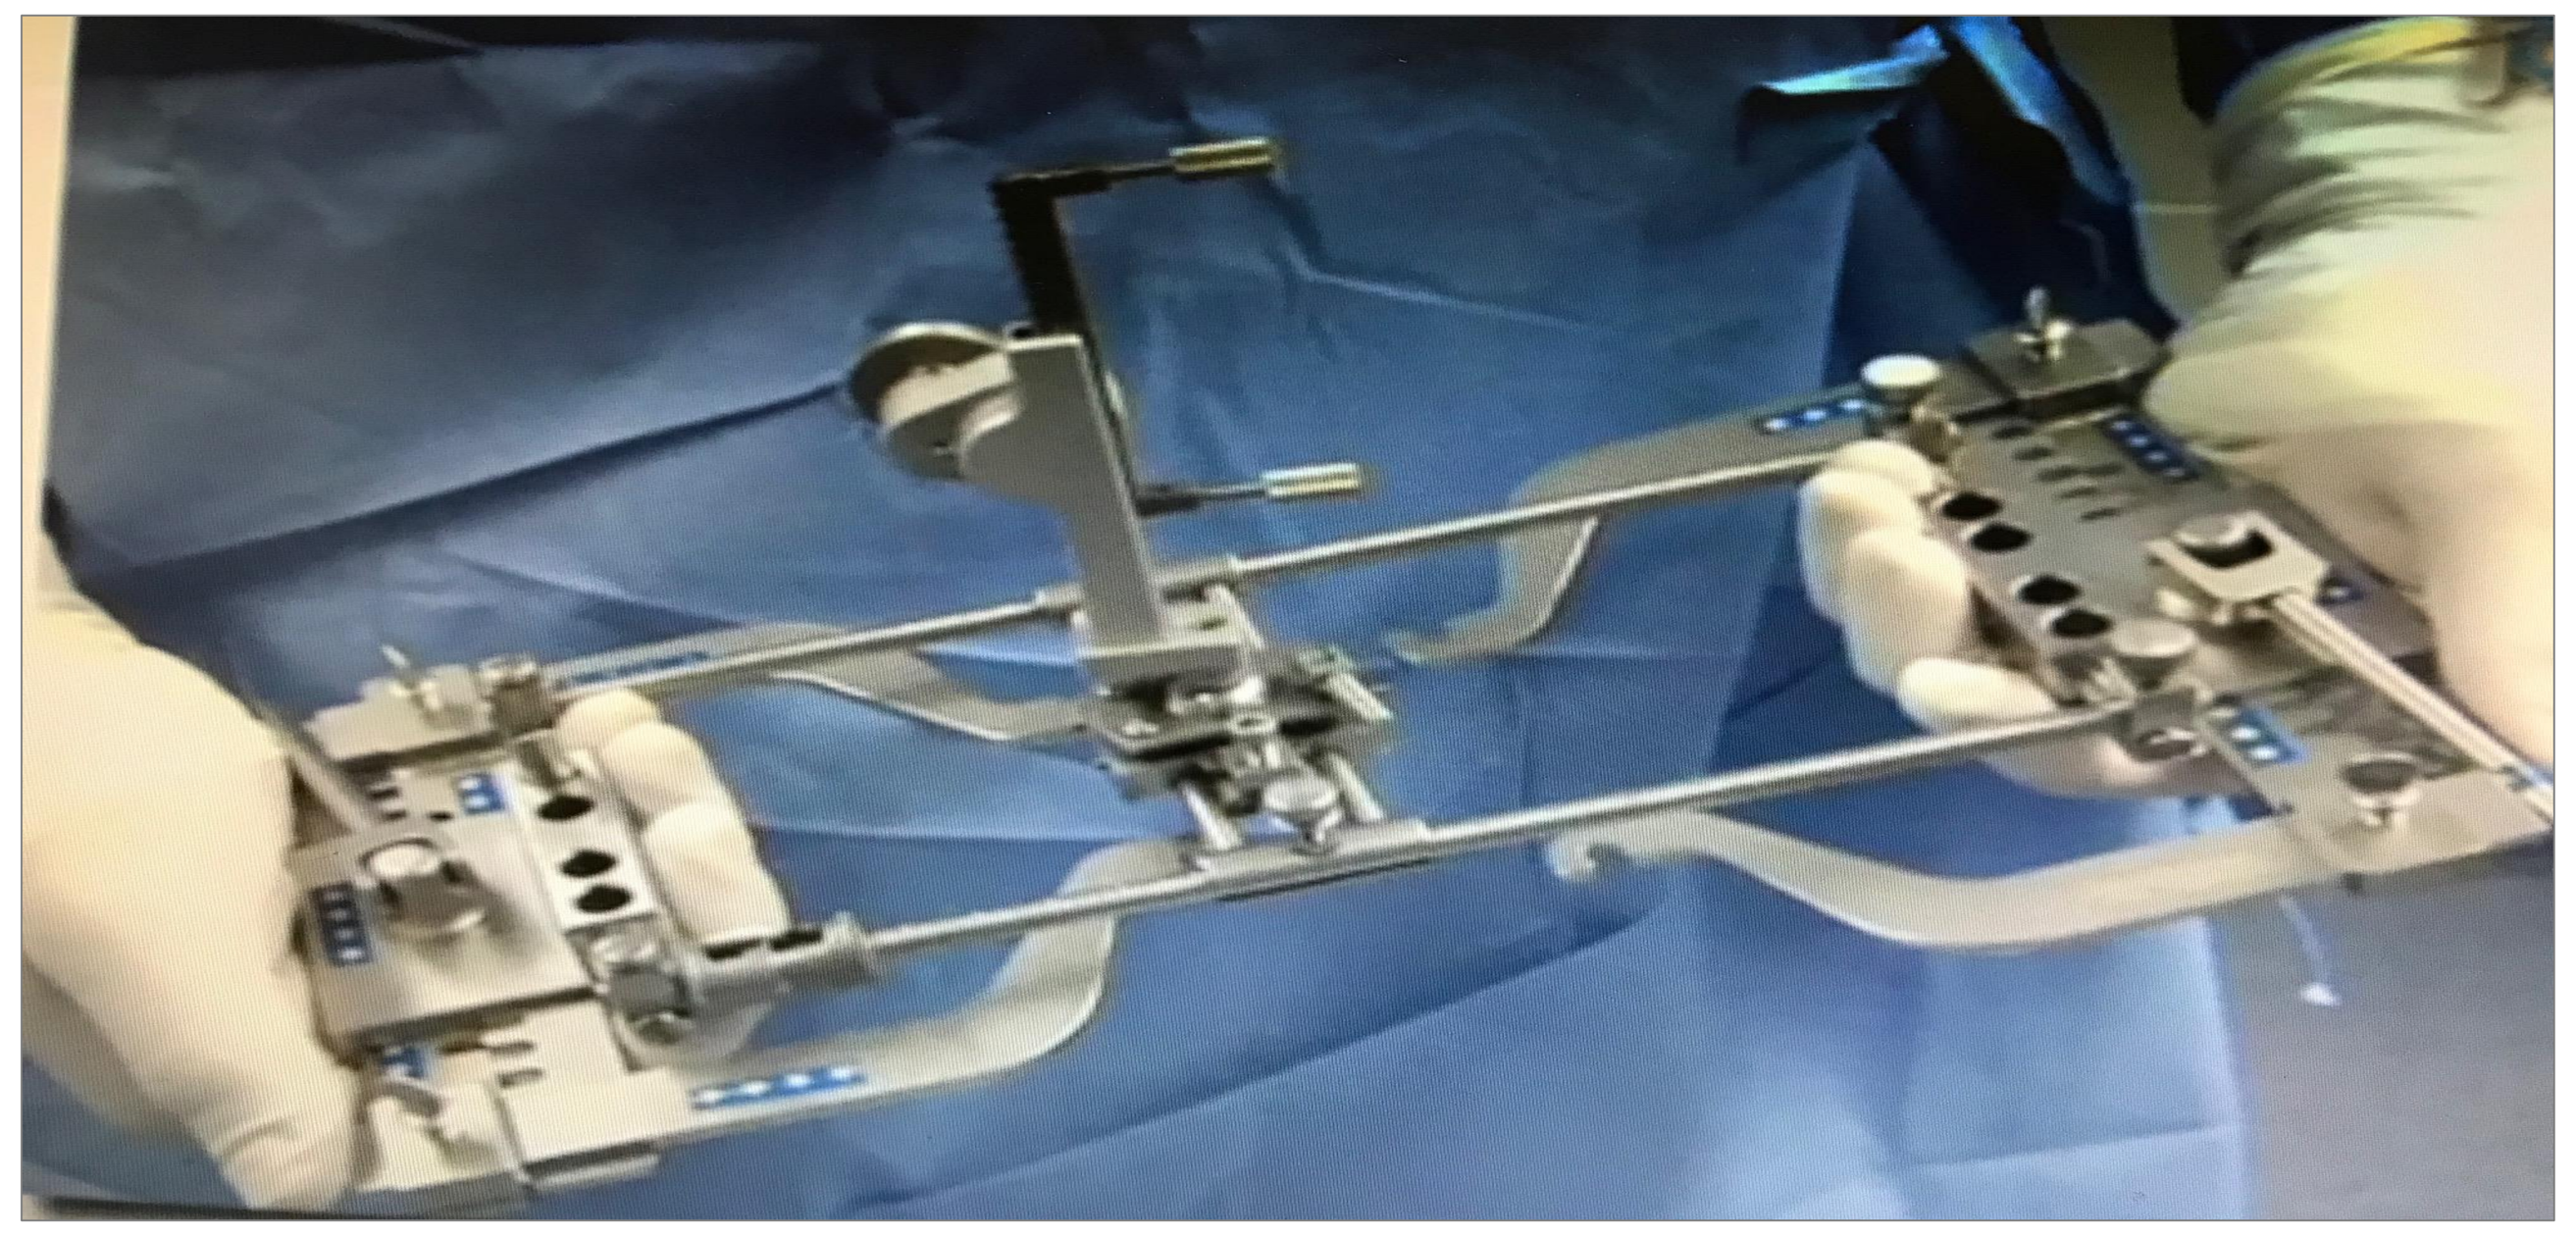

SITE OF ADMINISTRATION AND DOSING

Transplantation will target the posterior column of the spinal cord. Given that demyelinated axons in patients with TM is a significant contributor to disability, the ultimate goal is to administer Q-Cells® therapy to the spinal cord with demyelinated axons.

Initial transplantation studies will be performed in posterior columns unilaterally (Cohort 1; 2 million cells) followed by an increased number of cells in Cohort 2 (6 million cells) and Cohort 3 (12 million cells). Only transplantation foci below C7 cord level will be included in this study to avoid possible compromise of respiratory motor neuron pools.

Cohort	DOSE	[Cell] (cells/mL)	Volume (µL/site)
1	2	20 k	10
2	6	40 k	15
3	12	60 k	20

Acknowledgements

- Funding and support for this trial has been provided via the UT Southwestern CONQUER Program, the Transverse Myelitis Association and Q Therapeutics.

Disclosures

- Dr. Greenberg has received consulting fees from Alexion, EMD Serono and Novartis. He has received grant funding form NIH, NMSS, PCORI, Guthy Jackson Foundation, Transverse Myelitis Association, Chugai, Medimmune and Medday. He is an unpaid board member of the Transverse Myelitis Association